ATP Bioluminescence: A Tool to Measure Clinical Efficacy of Topical Oral Anti-microbials

N. Ramji*, R. Donovan-Brand, W. Buchanan, S. Hunter-Rinderle
P&G, Mason, OH, USA

4042

ABSTRACT

Objective: ATP bioluminescence assay is routinely used for microbiology quality assurance purposes. It provides rapid quantification of bacteria in biological samples. The current study uses this system to quantify viable bacteria in gingival crevicular fluid (GCF) using the Industrial Microbial Control (IMC) kit (Celsis Inc). **Methods:** The test procedure starts with the selective extraction of non-microbial (somatic) ATP by a specific somatic cell extractant (LuminEX(S)). The extractant makes the somatic cell membrane permeable for molecules like ATP. Microbial cells are not affected, because they have cell walls. A LuminASE hydrolyzes extracted somatic cell ATP. Microbial ATP in the sample is then extracted with LuminEX(B) and measured using a Celsis Advanced Luminometer after the addition of LuminATE (PM) which contains Luciferase and Luciferin. The Relative Light Units (RLU) produced correlates with the amount of bacterial ATP in the sample. A pilot study with 10 subjects was designed to validate the application of this technique in measuring clinical efficacy of antimicrobials. Changes in ATP bioluminescence and colony-forming units (cfu) of gram-negative anaerobes (GNA) from GCF samples were measured after 0.12% chlorhexidine (CHX) rinse. Overnight samples were collected from the subjects after three 15 mL CHX rinses for 30 seconds. Water was used as a control. **Results:** Analysis using paired-t test showed statistically significant reductions in bioluminescence ($p=0.009$) and cfu/mL ($p=0.002$) for the sub-gingival microorganisms after CHX treatment as compared to water control. The mean difference (Water minus Peridex) was 5063 RLU and 2.29x106 cfu/mL. **Conclusions: ATP bioluminescence assay could be effectively used as a pharmacodynamic tool to assess the clinical efficacy of dental products with antimicrobial activity.**

INTRODUCTION

Bacterial metabolism requires ATP and this can be used as a measure of viable bacteria in biological samples. ATP bioluminescence is a sensitive technique, which detects bacteria by measuring light emitted when their ATP reacts with firefly luciferase and luciferin. Standard microbiological techniques for culturing bacteria from samples take a minimum of 5 days to complete. ATP bioluminescence assay reduces this testing time to 24 hours. The test procedure uses Industrial Microbial Control (IMC) kit (from Celsis Inc.) which selectively extracts non-microbial (somatic) ATP by a specific somatic cell extractant LuminEX(S). The extractant makes the somatic cell membrane permeable for molecules like ATP, which leak out. Microbial cells were not affected. Extracted non-microbial ATP is next hydrolyzed using LuminASE(S). Microbial ATP is measured in a Celsis Luminometer after addition of LuminEX(B) and LuminATE(PM).

The amount of light produced correlates with amount of ATP in the sample, shown as Relative Light Units (RLU).

The light measurement is based on the following reaction:

	Mg^{++}	
Luciferin+ Luciferase + ATP	→	(Luciferin-Luciferase-AMP) + Pyrophosphate
	O_2	
(Luciferin-Luciferase-AMP)	→	Oxyluciferin + Luciferase + CO_2 +AMP + Light

The purpose of the current study was to quantify viable bacteria from GCF using the IMC kit and to evaluate the subgingival germ kill efficacy of a broad spectrum antimicrobial (often used as positive control) like chlorhexidine gluconate (Peridex) using ATP measurements and plating techniques.

MATERIALS AND METHODS

Study Design: This was an exploratory team test where a total of 10 subjects were evaluated for sub-gingival benefit after treatment with chlorhexidine gluconate. Prior to the study, subjects were charted to identify the sites for sample collection. Subjects were acclimatized on Crest Cavity Protection (CCP) toothpaste for 3 days and the morning of sample collection they came in un-brushed. The first day subjects brushed with CCP for 1 minute followed by rinsing with 15mL water for 15 seconds. As a part of the water treatment, subjects rinsed with 15mL water for 30 seconds in the morning, afternoon and evening and abstained from eating or drinking for 30 minutes after each treatment. On the second day, subjects reported to the clinic unbrushed and GCF samples were collected to evaluate the efficacy of the water treatments on sub-gingival microorganisms. After sample collection, subjects once again brushed with CCP and rinsed with 15mL water for 15 seconds. This was followed by rinsing with 15 mL 0.2% chlorhexidine gluconate (CHX) for 30 seconds in the morning, afternoon and evening. On the third day subjects once again reported to the clinic unbrushed and the GCF samples were collected to evaluate the efficacy of the chlorhexidine treatments on sub-gingival microorganisms.

Sample collection: Saliva and supra-gingival plaque were removed from the area around the GCF test sites. GCF sample was collected by inserting a strip of Periopaper (ORA Flow Inc., Babylon, NY) into the pocket of the sampling site in such a fashion that the apical end of the strip touched the deepest part of the midinterdental area. The strip was held for 30 seconds and the GCF collected was immediately measured using a Periotron (Periotron 8000). Samples were collected from two sites, the two periostrips were pooled into a sample vial containing 1 milliliter Reduced Transport Fluid (RTF) and placed in ice. The volume of sample collected, as determined by the Periotron, was used for normalization of data obtained from ATP and germ kill analysis.

Sample processing: A 1:100 and 1:1000 dilution of the GCF sample was cultured anaerobically on Enriched Tryptic Soya Agar containing Nalidixic acid and Vancomycin (ETSA-NV) for total gram negative anaerobes while the undiluted sample was analyzed for ATP bioluminescence.The colony counts on ETSA-NV were reported as colony forming units per milliliter (cfu/mL).

ATP studies: Sample was vortexed gently for 15 seconds. 75 µL of this was added to 75 µL of LuminEX(S) and 3 µL of LuminASE(S). This was gently vortexed to mix and incubated for 45 minutes at room temperature. 100 µL of the above was pipetted into a cuvette which was placed in the Luminometer. The Luminometer automatically injected 100 µL of LuminEX(B), extracted for 10 seconds and then injected 100 µL of LuminATE(PM). The result was recorded in RLU. A control or non-incubated sample was measured in the same way.

RESULTS

There was a significant positive correlation between the ATP data and germ kill for sub-gingival microorganisms after treatment with chlorhexidine gluconate treatment (CHX). Analysis using paired-t test showed statistically significant reductions in bioluminescence ($p=0.009$) and cfu/mL ($p=0.002$) for the sub-gingival microorganisms after CHX treatment as compared to water control. The mean difference (Water minus Peridex) was 5063 RLU and 2.29x106 cfu/mL. The was an exploratory team test and needs to be followed by a controlled clinical involving a strong/broad spectrum antimicrobial, as a part of the validation study (Table 1.).

Research presented at the 80th General Session of the IADR, March 6-9, 2002

P&G

4042

ATP Bioluminescence: A Tool to Measure Clinical Efficacy of Topical Oral Anti-microbials

N. Ramji*, R. Donovan-Brand, W. Buchanan, S. Hunter-Rinderle
P&G, Mason, OH, USA

Pg. 2

Figure 1. ATP bioluminescence (per subject analysis)

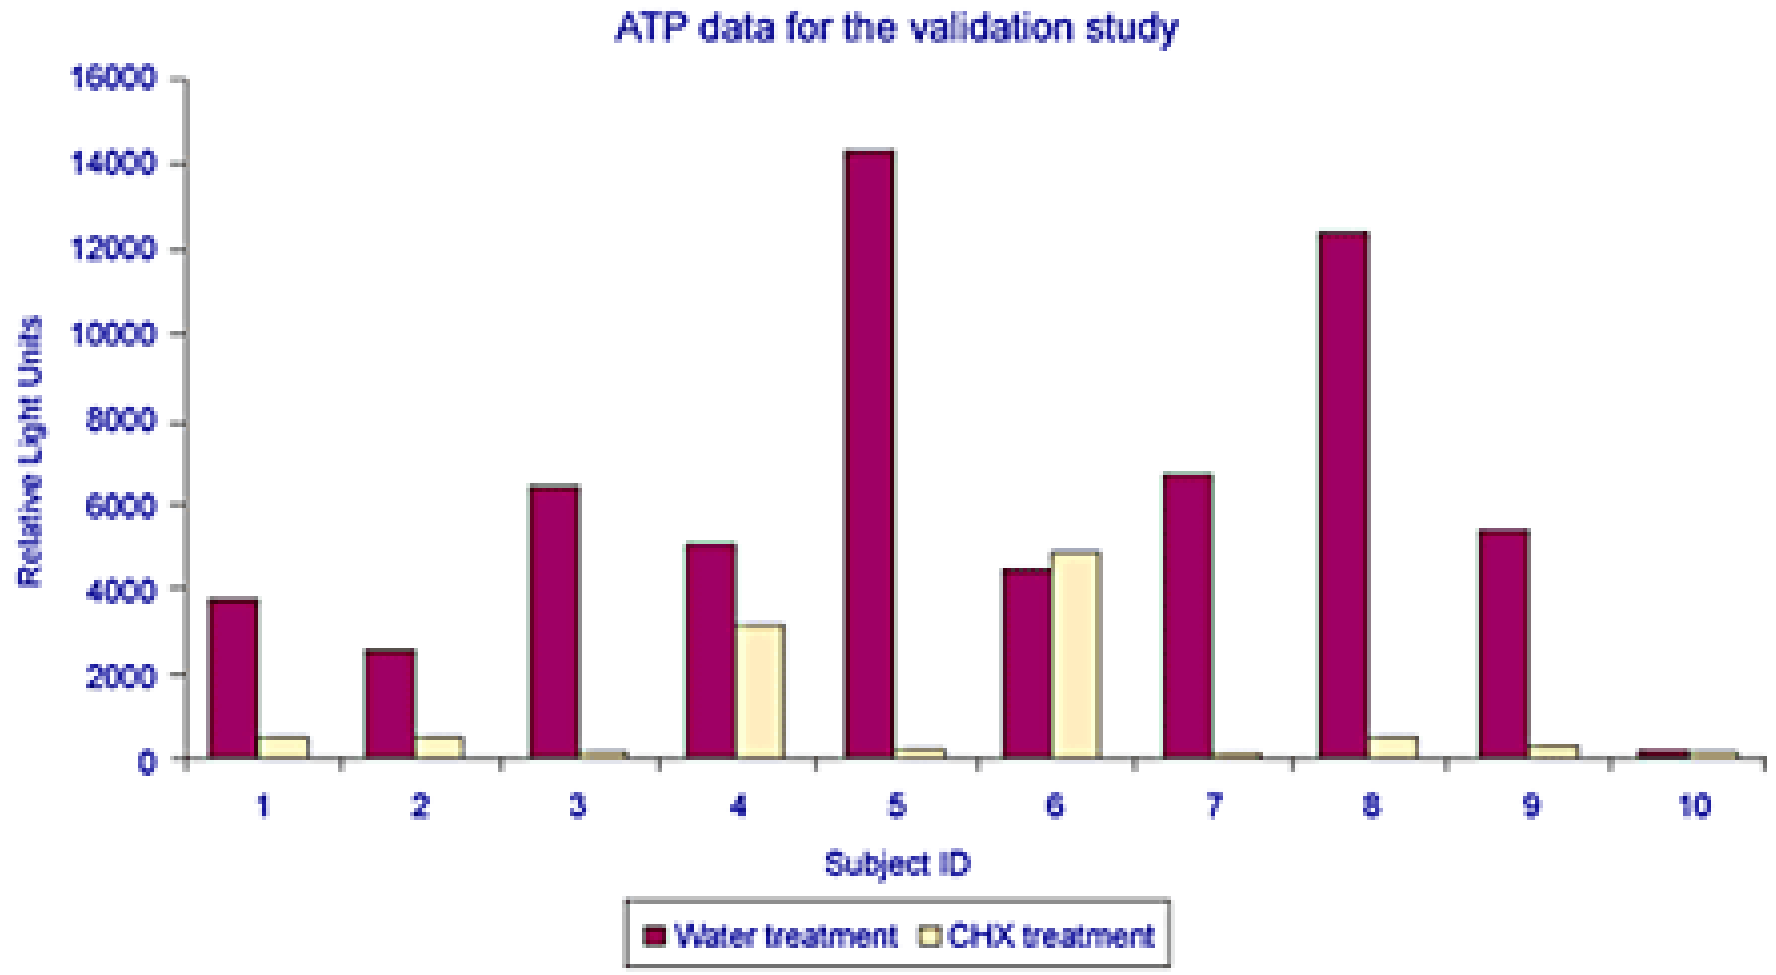
ATP data for the validation study
Relative Light Units
16000
14000
12000
10000
8000
6000
4000
2000
0
1
2
3
4
5
6
7
8
9
10
Subject ID
Water treatment
CHX treatment

Figure 2. Germ kill of GNAs on ETSA-NV (per subject analysis)

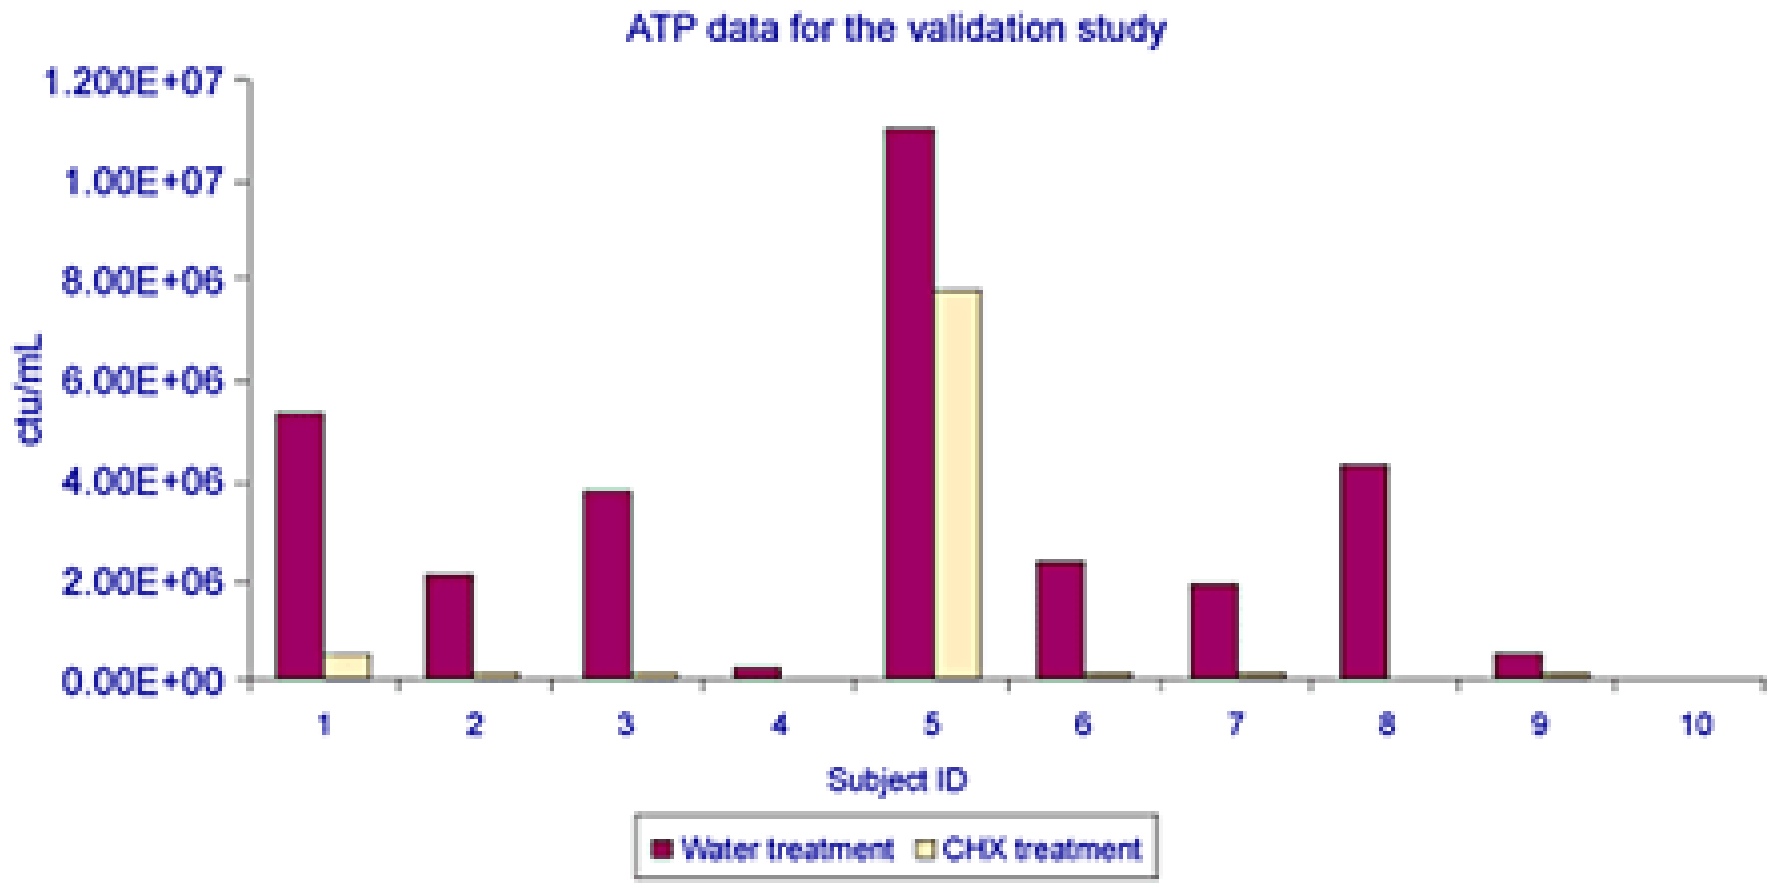
ATP data for the validation study
cfu/mL
1.200E+07
1.00E+07
8.00E+06
6.00E+06
4.00E+06
2.00E+06
0.00E+00
1
2
3
4
5
6
7
8
9
10
Subject ID
Water treatment
CHX treatment

FIGURES AND TABLES

Table 1. ATP and Germ kill analysis (average data)

Treatment	ATP data (Avg. RLU/µL)	Δ RLU/µL	P-value
Water	6097.9		
CHX	1034.8	-5063.1	0.009
Treatment	**Germ kill (Avg. cfu/µL)**	**Δ cfu/µL**	**P-value**
Water	3.20E+06		
CHX	8.80E+05	-2.30E+06	0.002

CONCLUSION

ATP bioluminescence may be effectively used to quantify metabolically active/ viable microorganisms in clinical samples and this test procedure may be used as a pharmacodynamic tool to evaluate the clinical efficacy of dental products with antimicrobial activity.

Acknowledgments: Susan Moffa (Customer Service Manager-Celsis Inc.), Brandon Peterson (Xavier University –Ohio).

Research presented at the 80th General Session of the IADR, March 6-9, 2002